A glycomacropeptide based protein substitute helps promote stable blood phenylalanine and branched chain amino acids in patients with phenylketonuria

***Robert M Browne*[1], Rachel Skeath[2], Paula Hallam[2], Melanie Hill[3], Carla Fitzachary[4], Heidi Chan[1,4], Joanna Gribben[4], Arlene Slabbert[4], Charlotte Ellerton[5], Francine Freedman[5], Kit Kaalund Hansen[5], Ide Herlihy[5], Karen van Wyk[6], Victoria Bittle[7], Emma Cameron[7], Gary P Hubbard[1], Rebecca J Stratton[1]**

[1] Medical Affairs, Nutricia Ltd, UK; [2] Great Ormond Street Hospital for Children NHS FT, UK; [3] Sheffield Teaching Hospitals NHS FT, UK; [4] Guy's & St Thomas' NHS FT, UK; [5] University College London Hospitals NHS FT, UK; [6] Manchester University NHS FT, UK; [7] Bristol University Hospitals NHS FT, UK

INTRODUCTION

Glycomacropeptide (GMP) based protein substitutes offer a promising alternative to 100% amino acid (AA) based protein substitutes for the dietary management of Phenylketonuria (PKU), due largely to the improved palatability of GMP. GMP-based protein substitutes contain the whey peptide GMP, alongside additional free AAs in order to create a balanced AA profile, often in addition to fats, carbohydrates, vitamins and minerals. However to date there is relatively limited data regarding how the AA profiles of currently available GMP-based protein substitutes may influence the blood AA profiles of patients with PKU.

The aim of this study was to evaluate the blood AA profile of patients introducing PhenylAde® GMP Drink Mix (Nutricia) into their diet for 28 days, with particular focus on Phenylalanine (Phe), Tyrosine (Tyr) and the Branched Chain Amino Acids (BCAAs): Isoleucine (Ile), Leucine (Leu) & Valine (Val).

METHODS

Twelve patients with PKU (see table 1) were recruited across 6 specialist hospitals in the UK. Patients undertook a 3 day baseline period during which they continued with their usual AA-based protein substitute(s), before introducing the study product (PhenylAde® GMP Drink Mix, 33.3g sachets, 10g Protein Equivalent), in an amount determined by their metabolic Dietitian for a 28 day intervention period.

Table 1. Patient Demographics (n=12)

Characteristic	Result
Mean Age (yrs)	28 [Range 5-50]
Sex (Female : Male)	9 : 3
Paediatric <16yrs : Adults ≥16yrs	4 : 8
Mean weight (kg)	59.5 (SD 27.3)
Mean height (m)	1.56 (SD 0.21)

The study product could either wholly or partially replace the patient's current AA-based protein substitute(s) and patients were advised to reduce the amount of Phe they consumed from food by an amount approximate to the residual Phe in the study product (15.3mg Phe/10g PE), to the nearest 25mg. Intake of the study product was self reported by the patient or parent/carer daily in order enable the calculation of average prescription compliance.

Fasting dried blood spots were collected on the morning after the baseline period, and days 7 and 28 of the intervention period, and blood AAs were analysed via High Performance Liquid Chromatography (Genova Diagnostics). The amino acids of focus were Phe, Tyr, Ile, Leu and Val. The analysis also assessed concentrations of Trp, His, Thr, Ala, Arg, Tau, Asp, Asn, Gln, Cys, Met, Ser, Gly, Orn, and Cit.

RESULTS

The mean prescription of the study product was 21.6g PE/d (range 10-60; SD 13.4), which was estimated to provide a mean of 34% of calculated total protein requirements (range 18-81; SD 17).

On average, the study product contributed 39.1% (range 22-100; SD 21.1) of protein equivalents prescribed from protein substitutes, with the remainder from amino acid supplements. Compliance to the study product was excellent (96%; SD 1.6).

Blood Phenylalanine

Blood Phe concentrations remained stable throughout the study period (Figure 1) with repeated measures ANOVA showing no significant changes over time (p=0.513 *NS*).

Blood Tyrosine

Blood tyrosine concentrations increased significantly over the study period (Figure 1). with repeated measures ANOVA showing a significant difference over time (p=0.02), although this did not result in a significant change in Phe:Tyr ratio (Baseline = 8.5 [SD 4.0], Day 7 = 7.1 [SD 3.6], Day 28 = 8.2 [SD 4.8]; p=0.24).

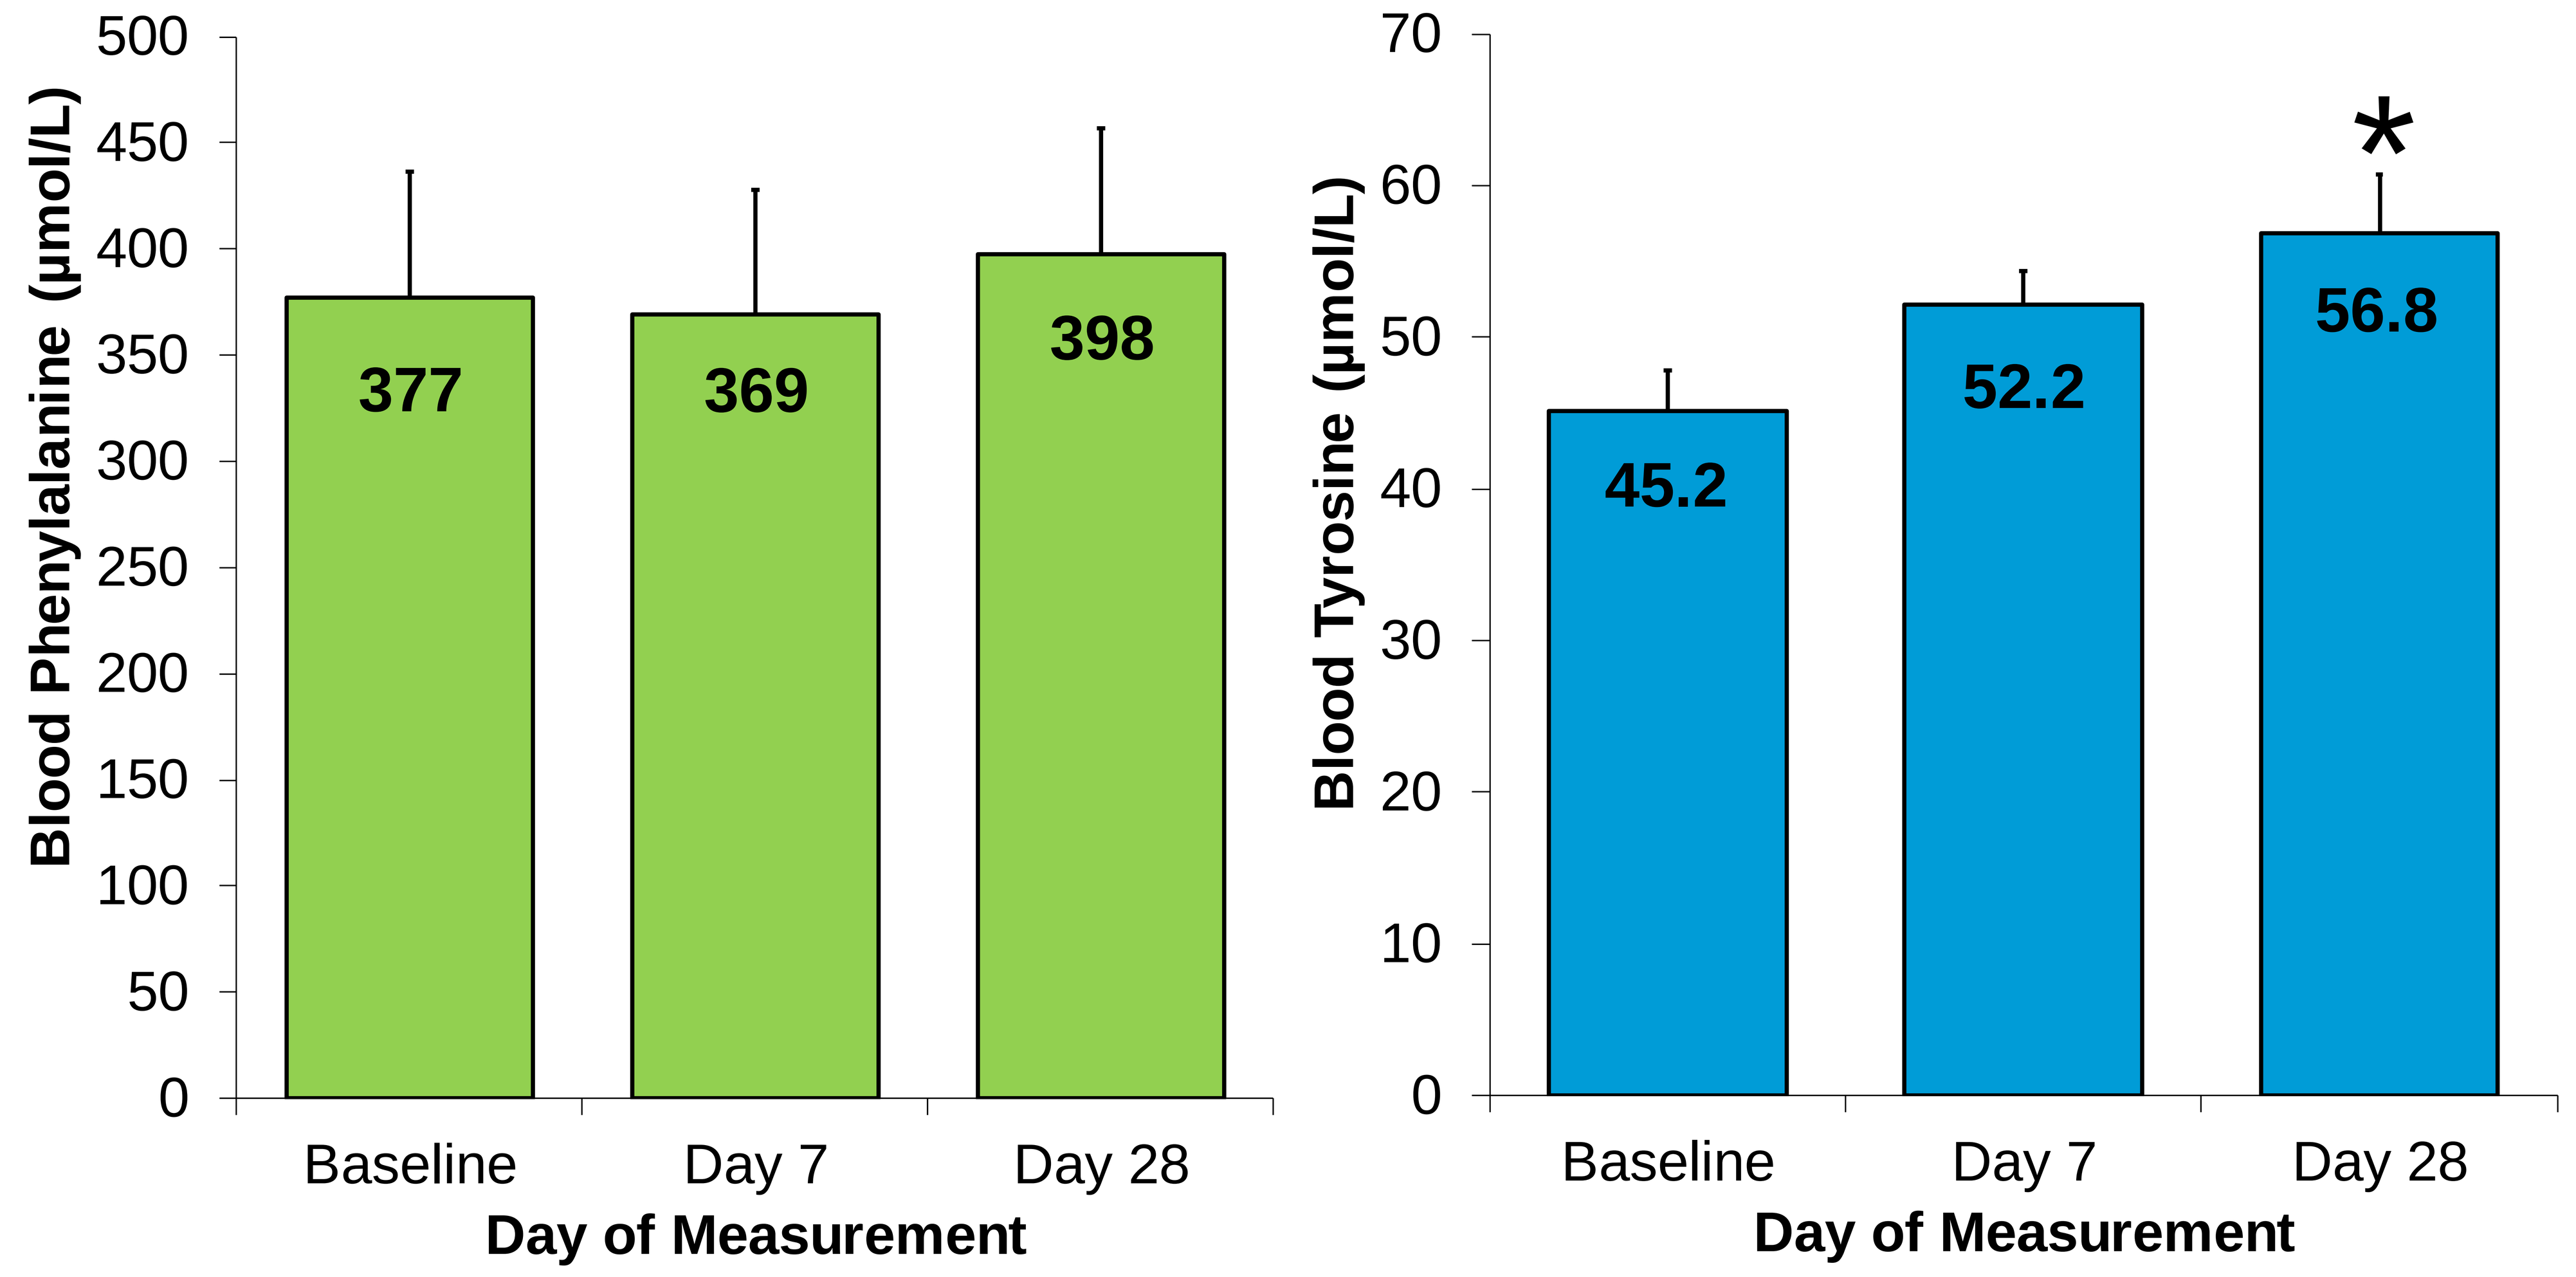
Blood Phenylalanine (µmol/L)
0
50
100
150
200
250
300
350
400
450
500
377
369
398
Baseline
Day 7
Day 28
Day of Measurement
Blood Tyrosine (µmol/L)
0
10
20
30
40
50
60
70
45.2
52.2
56.8
*
Baseline
Day 7
Day 28
Day of Measurement

Figure 1. Mean dried blood spot amino acid results for Phenylalanine and Tyrosine (µmol/L). Error bars: SEM. * RMANOVA illustrated significant change over study (p<0.05).

Branched Chain Amino Acids (BCAAs)

All BCAAs remained stable over the study period (p>0.05) and the ratios of Ile:Leu:Val did not significantly change (p>0.05), remaining at approximately 1 : 2 : 3.8 (Table 2).

Table 2. Mean (SD) dried blood spot BCAA results (µmol/L) and BCAA ratios.
[ab] BCAA ratios analysed statistically as: [a] Ile:Leu and; [b]Ile:Val but presented as Ile:Leu:Val for clarity.

Amino Acid(s)	Baseline	Day 7	Day 28	p-value
Ile	41 (11)	39 (8)	43 (13)	0.57
Leu	83 (19)	79 (18)	83 (22)	0.56
Val	158 (42)	146 (33)	153 (42)	0.38
Ile : Leu : Val	1 : 2.07 : 3.89	1 : 2.04 : 3.81	1 : 2.03 : 3.72	0.91[a] / 0.76[b]

These positive results remained consistent for the patients who consumed ≥50% of their protein requirements from the study product (n=2), though no statistical analyses could be performed for this subset. Overall, during the intervention period, 95% of results for all 20 amino acids analysed (excluding Phe) were found to be within 95% population reference ranges.

CONCLUSIONS

PhenylAde® GMP Drink Mix resulted in no significant changes in blood AAs over the 28 day intervention period, with the exception of a significant improvement in tyrosine levels. BCAA ratios remained within recommended ranges. These results demonstrate that PhenylAde® GMP Drink Mix is safe and effective for the dietary management of PKU in adults and children.